Comparison of uniresponse and multiresponse approaches of PopDes to optimize sampling times for drug-drug interaction studies: Application to a Servier compound.

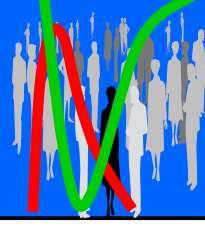

Marylore Chenel[1], Kayode Ogungbenro[2], François Bouzom[1]

[1] Servier, France, [2] School of Pharmacy, University of Manchester, UK

INTRODUCTION

A Drug-Drug Interaction (DDI) study was planned to evaluate the potential inhibitory effect of a phase I Servier compound (called SX) on a reference CYP3A4 substrate, Midazolam (MDZ).

PBPK models allow to simulate PK profiles of SX & MDZ when administered separately but also together (DDI) from *in vitro* parameters. Multiresponse design as implemented in PopDes allows to estimate joint optimal sampling times for 2 drugs. Therefore, we decided to design at best (minimum sampling time number) the clinical trial without any *in vivo* data using PBPK predictions, population PK modelling and multiresponse optimal design.

Objectives:

1) To compare designs obtained by uniresponse & multiresponse design approaches of PopDes.

2) On real data, to compare MDZ apparent clearance (CL/F) estimates obtained by population PK modelling using either a full empirical design or the optimal design obtained with the multiresponse design approach.

1- METHODS

1.1-Study Design and Data:

➢ 100 PK profiles using PBPK models for both drugs taking into account the potential DDI were simulated from *in vitro* parameters.

➢ The simulated design was the one planned for the clinical trial: 12 subjects receiving SX dose b.i.d. over 5 days, and the 5th day, a single dose of MDZ given 2h after 1rst daily SX dose (Fig. 1).

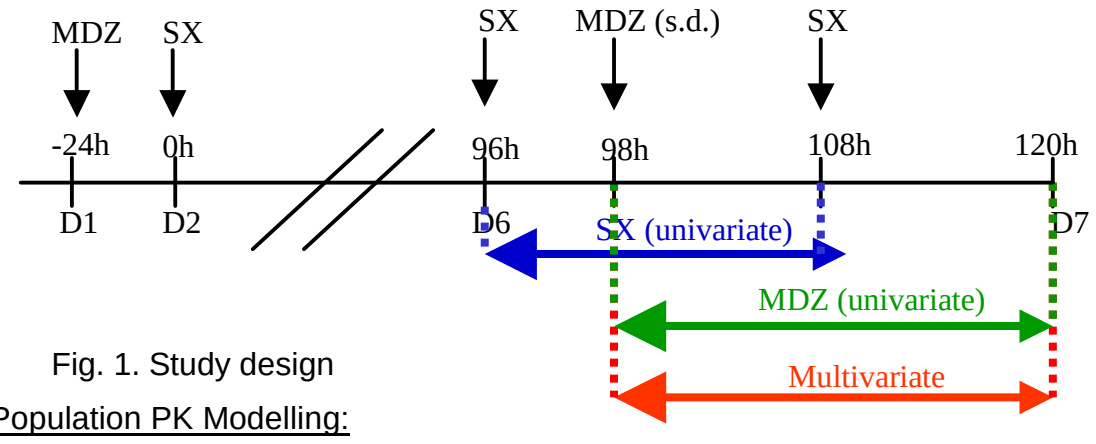

MDZ
SX
SX
MDZ (s.d.)
SX
-24h
0h
96h
98h
108h
120h
D1
D2
D6
D7
SX (univariate)
MDZ (univariate)
Multivariate

Fig. 1. Study design

1.2-Population PK Modelling:

➢ Simulated SX & MDZ PK data were fitted using NONMEM[1] V (FOCEI).

1.3-Sampling time optimization by uniresponse & multiresponse design approaches with PopDes:

➢ Design domain (Fig. 1) : - Uniresponse design (UOD) over 12h & 22h for SX & MDZ, respectively.

- Multiresponse design (MOD) over 22h for both compounds.

➢ A single group of 12 subjects was considered.

➢ Use of population parameters estimates (Local) / Criteria: D-optimality / Algorithm: Federov exchange[2].

1.4-Evaluation of designs obtained by uniresponse and multiresponse by simulations:

➢ Simulation & estimation of 1000 datasets with NONMEM (FOCEI).

➢ Comparison of RMSE and estimation accuracy (empirical RSE, mean RSE given by NONMEM & RSE given by PopDes) of CL/F for both drugs.

1.5- Design of the clinical trial:

Sampling times of the MOD were slided into an empirical full design (FD) with 11 sampling times.

1.6- Analysis of observed MDZ data and comparison of designs:

➢ Observed MDZ data were fitted using NONMEM V & MONOLIX 2[3] under 2 settings (alone or +SX) using either the empirical FD or the MOD.

➢ MDZ CL/F_{MDZ+SX} / $CL/F_{MDZ\ alone}$ ratios obtained with the FD and the MOD were compared.

2- RESULTS

2.1-Population PK modelling:

➢ SX simulated PK data were fitted by a 2-compartment model with a fixed Ka, intra-individual variability (IIV) on CL/F and on Vc/F, a correlation between CL/F & Vc/F and a combined error model.

➢ MDZ simulated PK data were fitted by a 2-compartment model with a zero-order absorption constant, IIV on all parameters, a correlation between CL/F & Vc/F and a combined error model.

2.2-Sampling time optimization using PopDes:

➢ Uniresponse Design: **SX = 4 sampling times [0h, 40min, 4h & 12h] after 96h**

MDZ = 5 sampling times [15min, 50min, 5h, 12h15min & 22h] after 98h

➢ Multiresponse Design: **SX & MDZ = 5 sampling times [15min, 1h, 5h30min, 10h30min & 22h] after 98h**

2.3-Evaluation of designs obtained by uniresponse and multiresponse by simulations :

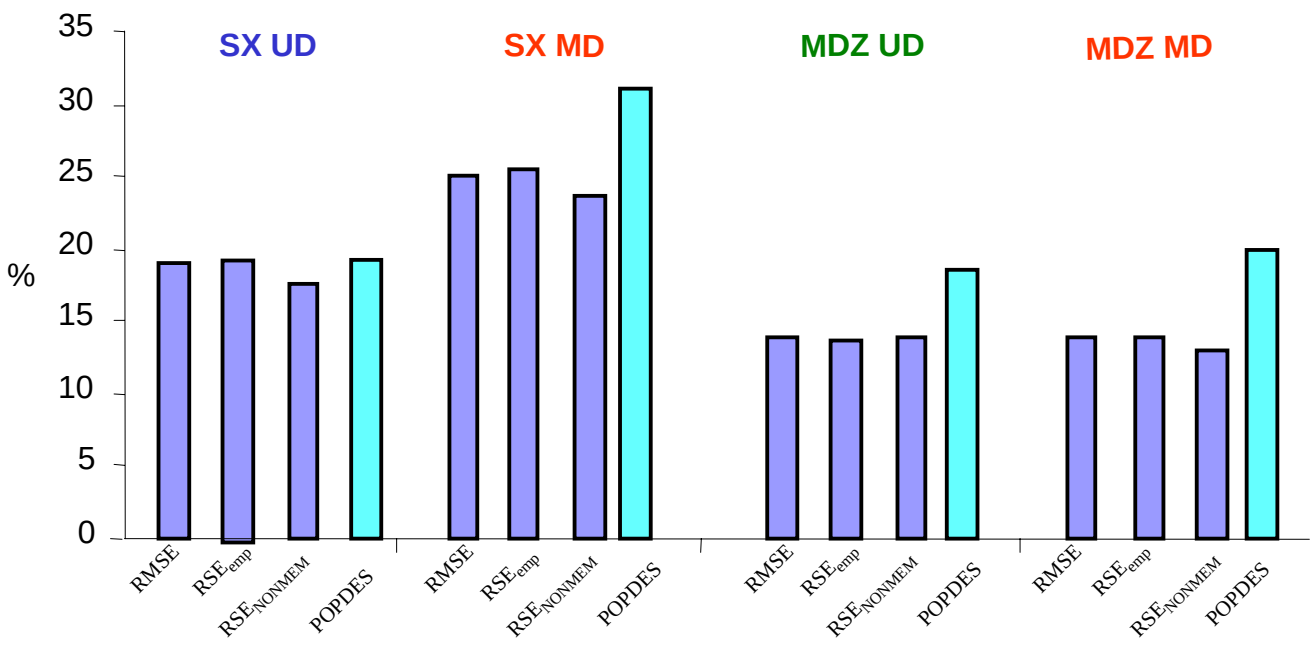

SX UD
SX MD
MDZ UD
MDZ MD
%
0
5
10
15
20
25
30
35
RMSE
RSEemp
RSENONMEM
POPDES

Fig. 2. CL/F RMSE and accuracy of estimation (RSE) for SX & MDZ using uniresponse designs (UOD) and multiresponse designs (MOD) .

2.4-Analysis of observed MDZ data and design comparison:

MDZ CL/F (L/h) (RSE%)	Design	MDZ alone	MDZ+SX	Ratio
NONMEM	FD	75 (12)	40 (10)	0.53
	MOD	101 (nd*)	50** (nd*)	0.49**
MONOLIX	FD	72 (12)	40 (11)	0.55
	MOD	81 (57)	45 (29)	0.55

nd* = not determined because covariance step aborted in NONMEM
** = minimization terminated with the error 134 in NONMEM

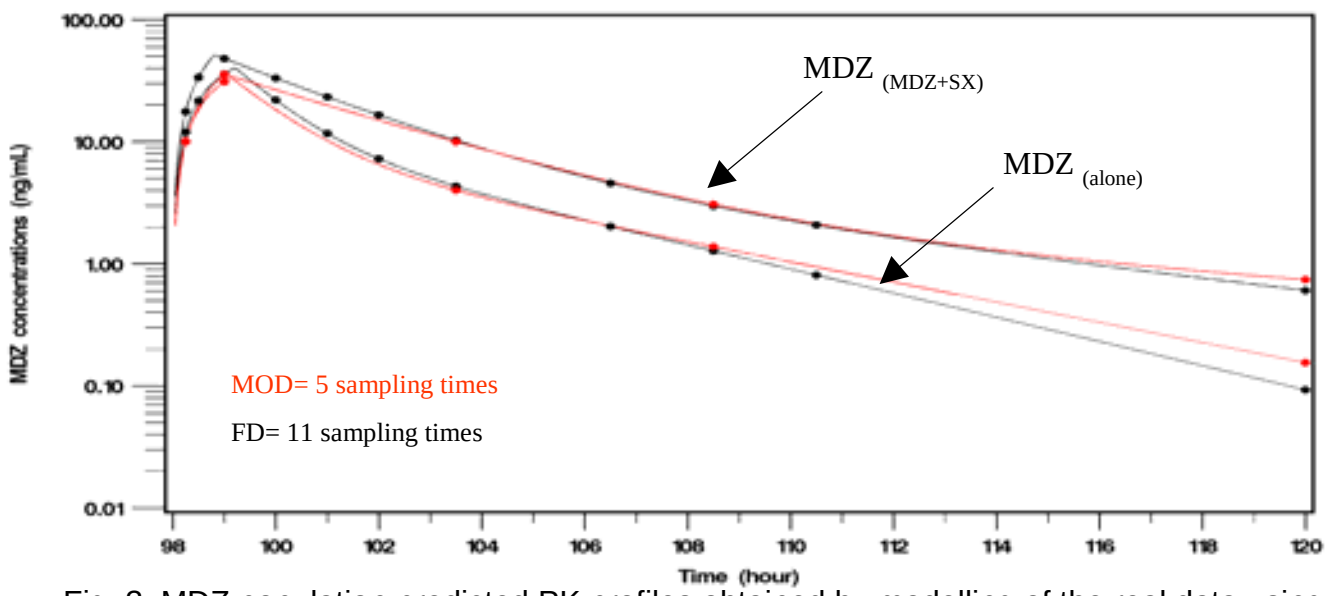

MDZ (MDZ+SX)
MDZ (alone)
MOD= 5 sampling times
FD= 11 sampling times
MDZ concentrations (ng/mL)
100.00
10.00
1.00
0.10
0.01
98
100
102
104
106
108
110
112
114
116
118
120
Time (hour)

Fig. 3. MDZ population predicted PK profiles obtained by modelling of the real data using either the full design (FD) or the multiresponse optimal design (MOD) when the MDZ was administered alone or with SX.

3- DISCUSSION

3.1- Design of the clinical trial and evaluation

➢ PBPK models allowed to predict from *in vitro* data PK profiles of SX and MDZ when they are co-administered.

➢ Multiresponse approach as implemented in PopDes allowed to optimize joint sampling times for 2 drugs.

➢ Evaluation of designs by simulations & estimations showed that CL/F was well estimated for MDZ (< 20%) and correctly for SX with the both design approaches (< 30%) (Fig. 2).

➢ Empirical RSE, mean RSE obtained from NONMEM RSEs as well as RSE given by PopDes were in the same range. The CL/F RMSE was also in the same range than RSEs showing that there was no bias (Fig. 2).

➢ The MOD with joint sampling times for the 2 drugs allowed to save 4 sampling times (9 *vs* 5) compared to the 2 Uniresponse Optimal Designs (therefore 12 subjects x 4 sampling times = 48 samples and 48 samples x 2 drugs = 96 analysed samples).

3.2- Clinical trial results (analysis of real data)

➢ The interaction ratio predicted by the PBPK model was about 2 fold lower than the observed one.

➢ The predicted inter-individual variability as well as the residual error of the PBPK models were much lower then the ones observed (results not shown).

➢ NONMEM failed (error 134) to estimate population PK parameters with the MOD when the MDZ was co-administered with SX (maybe due to the unpredicted interaction) and was not able to give RSEs (covariance step aborted) when the MDZ was given alone with the MOD.

➢ MONOLIX was able to estimate population PK parameters with the FD as well as with the MOD in both cases (MDZ alone or co-administered with SX).

➢ NONMEM estimates were close to MONOLIX estimates and inhibition ratios were comparable leading to the same conclusion.

➢ The MOD (5 sampling times) allowed to predict the same inhibition ratio than the one obtained with the FD (11 sampling times).

CONCLUSION

Under the clinical constraints for these 2 population PK models, the multiresponse design approach with joint optimal sampling times allowed MDZ CL/F values to be well estimated in addition of PK information collection for SX, and allowed to save 4 sampling times compared to the uniresponse design approach. Thus, for the clinical trial, the optimal sampling times estimated by both approaches were slided into the full anticipated sampling time design.

This global approach including PBPK simulations, population PK modelling and multiresponse optimal design allowed without any *in vivo* data to design a clinical trial using sparse sampling able to detect a PK interaction between 2 co-administered drugs. Nevertheless, variability of PBPK models should be sometimes increased to be more realistic.

The clinical trial finished, real data analysis showed that the multiresponse optimal design allowed to give the same conclusion (a factor 2 decrease of the MDZ CL/F when co-administered with SX) than the full empirical design.

Acknowledgment to Pr. Leon Aarons, Pr. France Mentré and the Clinical PK department in Servier

[1]Beal & Sheiner, 1996, *NONMEM Users Guides, Version 5, NONMEM Project Group*, U.C.S.F. [2]Ogungbenro, Gueorguieva, Graham & Aarons, 2005, *Comp. Meth. Prog. Biomed.* [3]Lavielle & Mentré, 2007, *JPP*..